Incidence and severity of diarrhea with neratinib plus intensive loperamide prophylaxis in patients with HER2-positive early-stage breast cancer: interim analysis from the multicenter, open label, phase II CONTROL trial

Carlos H. Barcenas,[1] Emad Ibrahim,[2] Mary Wilkinson,[3] Sara Hurvitz,[4] Nicholas Iannotti,[5] Andrew Kellum,[6] Yvonne Manalo,[7] Serena Wong,[8] Vincent Hansen,[9] Ricardo H. Alvarez,[10] Arlene Chan,[11] Ira Gore,[12] S. DiSean Kendall,[13] James L. Wade III,[14] Elizabeth Olek,[15] Daniel Hunt,[15] Pearl Fang,[15] Azita Ebtahaj,[15] Federico Ustaris,[15] Susan Moran,[15] Debu Tripathy[1]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Beaver Medical Group, L.P., Highland CA; [3]Inova Medical Group, Fairfax, VA; [4]UCLA Hematology/Oncology Clinical Research Unit, Los Angeles, CA; [5]Hematology Oncology Associates of the Treasure Coast, Port Saint Lucie, FL; [6]North Mississippi Medical Center Hematology and Oncology Services, Tupelo, MS; [7]Coastal Bend Cancer Center, Corpus Christi, TX; [8]Rutgers Cancer Institute of New Jersey, New Brunswick, NJ; [9]Utah Hematology Oncology, Ogden, UT; [10]Southeastern Regional Medical Center Inc., Neunan, GA; [11]Hollywood Private Hospital, Nedlands, WA, Australia; [12]Alabama Oncology, Bruno Cancer Center, Birmingham, AL; [13]Utah Cancer Specialists Research, Salt Lake City, UT; [14]Cancer Care Specialists, Decatur, IL; [15]Puma Biotechnology Inc., Los Angeles, CA

#P2-11-03

Background

- Neratinib (Puma Biotechnology Inc.) is an irreversible pan-HER tyrosine kinase inhibitor.[1]
 - Results from the randomized, Phase III ExteNET study demonstrated that a 1-year course of neratinib after trastuzumab-based adjuvant therapy reduced the risk of disease recurrence or death by 33% relative to placebo after 2 years' follow-up in women with HER2-positive early-stage breast cancer.[2]
 - Patient follow-up is ongoing; the final 5-year efficacy analysis is anticipated in 2017.
- Diarrhea is the main toxicity of neratinib and is common in the absence of proactive management.[3]
 - In the ExteNET trial, where antidiarrheal prophylaxis was not protocol mandated:
 - Median duration of grade ≥3 diarrhea was 5 days;
 - Neratinib dose reductions and dose holds due to diarrhea occurred in 26.4% and 33.9% of patients, respectively.[2]
 - As most diarrhea events occur early in the course of neratinib treatment, a structured (intensive) prophylactic regimen of loperamide given for 1–2 cycles has been introduced in all clinical trials of neratinib to better manage this toxicity.[3]
 - Recent preclinical studies suggest that multiple mechanisms may be involved in the pathogenesis of neratinib-induced diarrhea, including elements of secretory and inflammatory diarrhea.[4] In particular, in a rat model, inflammation was observed in the terminal ileum.[4]
- CONTROL is an international, open-label, phase II study designed to investigate the efficacy of 1–2 cycles of loperamide prophylaxis in the prevention of neratinib-associated diarrhea in patients with HER2-positive early-stage breast cancer.
 - The study has been expanded to include additional patient cohorts treated with agents targeting possible underlying mechanisms (e.g. anti-inflammatories, bile acid sequestrants) to determine if they can further reduce neratinib-associated diarrhea.
 - We report an interim analysis from the CONTROL study, and preliminary findings from the first investigational cohort testing budesonide, a locally acting corticosteroid used for inflammatory gastrointestinal conditions.

Methods

Study design

- CONTROL (PUMA-NER-6201) is an international, open-label, phase II study (Clinicaltrials.gov identifier NCT02400476).

Figure 1. Study design and flowchart

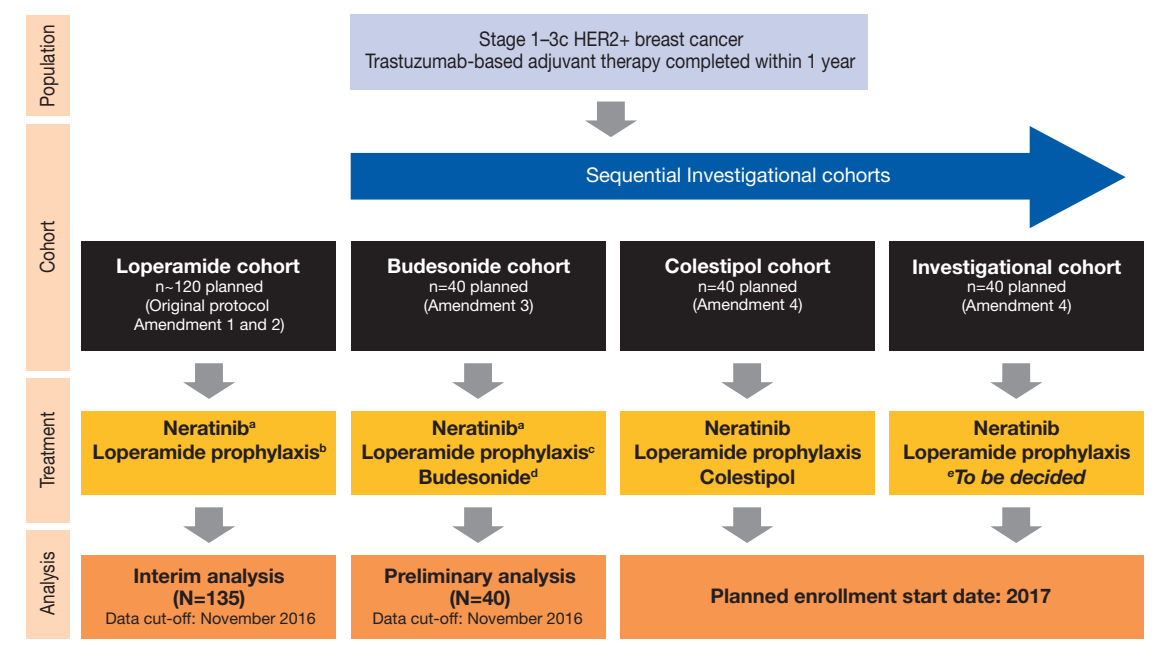
Population
Cohort
Treatment
Analysis
Stage 1–3c HER2+ breast cancer
Trastuzumab-based adjuvant therapy completed within 1 year
Sequential Investigational cohorts
Loperamide cohort
n~120 planned
(Original protocol Amendment 1 and 2)
Budesonide cohort
n=40 planned
(Amendment 3)
Colestipol cohort
n=40 planned
(Amendment 4)
Investigational cohort
n=40 planned
(Amendment 4)
Neratinib[a]
Loperamide prophylaxis[b]
Neratinib[a]
Loperamide prophylaxis[c]
Budesonide[d]
Neratinib
Loperamide prophylaxis
Colestipol
Neratinib
Loperamide prophylaxis
[e]To be decided
Interim analysis
(N=135)
Data cut-off: November 2016
Preliminary analysis
(N=40)
Data cut-off: November 2016
Planned enrollment start date: 2017

[a]Neratinib 240 mg once daily for 13 cycles.
[b]Loperamide prophylaxis for 2 cycles: 4 mg initial dose, then 2 mg q4h days 1–3 (i.e. 12 mg/day), then 2 mg q6–8h 4–56 (i.e. 6–8 mg/day) *(original schedule)* or loperamide 4 mg initial dose, then 4 mg tid days 1–14 (i.e. 12 mg/day), then 4 mg bid days 15–56 (i.e. 8 mg/day) *(modified schedule)*.
[c]Loperamide prophylaxis for 2 cycles: 4 mg initial dose, then 4 mg tid days 1–14 (i.e. 12 mg/day), then 4 mg bid days 15–56 (i.e. 8 mg/day) *(modified schedule)*.
[d]Budesonide 9 mg once daily (extended-release tablets) 1 cycle.
[e]Agent selected for this cohort will be based on the review of ongoing preclinical investigations.
One cycle = 28 days; bid, twice daily; tid, 3-times daily; q = every.

Patient population

- Adults ≥18 years of age.
- Histologically confirmed stage 1–3c breast cancer.
- Documented HER2 overexpression or amplification determined locally.
- Completed trastuzumab-based adjuvant therapy, or experienced side effects resulting in early discontinuation, with last trastuzumab dose given >2 weeks and ≤1 year prior to enrollment.

Study treatments

- Details of treatment schedules are presented in Figure 1.
- Eligible patients were to receive:
 - Oral neratinib 240 mg/day for 1 year.
 - Oral loperamide prophylaxis for 2 cycles according to one of two schedules:
 1. Original schedule (first protocol).
 2. Modified schedule to simplify dosing and improve efficacy (protocol amendments 1–3).
 - Oral budesonide for 1 cycle (protocol amendment 3).
- Loperamide (≤16 mg/day) was given as needed after day 56.
- Treatment-emergent diarrhea was managed with dietetic measures and additional pharmacological treatments depending on grade (i.e. diphenoxylate plus atropine, octreotide, IV fluids, antibiotics).

Assessments

- Clinic visits were scheduled on day 1 of cycles 1, 2, 3, 4, 7 and 10, and treatment end.
- Patients were followed for 28 days after the last dose of neratinib.
- Adverse events were graded according to NCI-CTCAE (version 4.0).

Endpoints

- *Primary endpoint:* incidence of grade ≥3 diarrhea during treatment with neratinib.
- *Secondary endpoints:* frequency distribution of maximum-grade diarrhea; incidence and severity of diarrhea by loperamide exposure; serious adverse events; adverse events of interest.
- *Exploratory endpoints*: patient-reported outcomes; biomarkers for disease recurrence.

Statistical methods

- All analyses were descriptive and were performed in the safety population, defined as all patients who received ≥1 dose of neratinib.
- A protocol-defined interim analysis was performed when approximately 120 patients from the loperamide cohort had completed 2 cycles of neratinib plus loperamide prophylaxis. A preliminary analysis of the budesonide cohort was also performed.
- The ExteNET trial (neratinib arm), which included an analogous patient population but no protocol-mandated antidiarrheal prophylaxis,[2] was used as a historical control.

Results

Table 1. Baseline characteristics

	CONTROL		ExteNET
Variable	Loperamide cohort (N=135)	Budesonide cohort (N=40)	Neratinib arm (N=1420)
Female, %	100	100	100
Median age (range), years	53 (30–86)	50 (29–78)	52 (25–83)
Tumor stage at diagnosis,[a] %			
I	28.9	22.5	9.8
IIA, B	54.8	42.5	42.0
IIIA, B, C	14.8	17.5	31.2
Hormone receptor status,[b] %			
Positive (ER and/or PR positive)	74.1	62.5	57.5
Negative (ER and PR negative)	25.2	25.0	42.5
Prior (neo)adjuvant therapy,[c] %			
Trastuzumab	100	87.5	100
Taxanes	95.6	90.0	90.1
Anthracycline	25.9	25.0	77.3
Pertuzumab	40.0	55.0	–
Median (range) duration of prior trastuzumab, months	11.5 (2.4–24.9)	11.0 (1.2–15.0)	11.5 (0.7–56.9)
Median (range) time since last trastuzumab dose, months	4.1 (0.1–19.8)	4.3 (0.7–11.7)	4.4 (0.2–30.9)

ER, estrogen receptor; PR, progesterone receptor.
[a]Unknown: loperamide cohort, n=2; budesonide cohort, n=7; ExteNET (neratinib arm), n=241.
[b]Missing: loperamide cohort, n=1; budesonide cohort, n=5.
[c]Budesonide cohort: no or missing (n=4).

- A total of 135 patients were included in the interim analysis of the loperamide cohort (original loperamide schedule, n=28; modified loperamide schedule, n=107).
- Forty patients were included in the preliminary analysis of the actively enrolling budesonide cohort (Figure 1).
- Baseline characteristics are presented in Table 1.

Treatment-emergent diarrhea

- Incidence of grade ≥3 diarrhea, the primary study endpoint, was 28.1% (95% CI 20.8–36.5%) with loperamide prophylaxis (loperamide cohort) and 15.0% (95% CI 5.7–29.8%) with loperamide prophylaxis plus budesonide (budesonide cohort) vs 39.9% without protocol-mandated loperamide prophylaxis in the ExteNET trial.
- A summary of treatment-emergent diarrhea is presented in Table 2.

Table 2. Characteristics of treatment-emergent diarrhea

	CONTROL				ExteNET[2]
	Loperamide cohort			Budesonide cohort	Neratinib arm
	Original schedule (n=28)	Modified schedule (n=107)	Loperamide Total (N=135)	Loperamide + budesonide (N=40)	Loperamide prn (N=1408)
Diarrhea, %					
Any grade	82.1	73.8	75.6	65.0	95.4
Grade 1	35.7	21.5	24.4	32.5	22.9
Grade 2	21.4	23.4	23.0	17.5	32.5
Grade 3[a]	25.0	29.0	28.1	15.0	39.8
Grade 4	0	0	0	0	0.1
Median cumulative duration, days					
Grade ≥2	5.0	4.0	4.0	3.0	10.0
Grade ≥3[b]	2.0	3.0	3.0	2.5	5.0
Median diarrhea episodes/patient					
Any grade	2	2	2	2	8
Grade ≥2	2	1	2	1	3
Grade ≥3[b]	1	1	1	1	2
Action taken, %					
Dose hold	7.1	12.1	11.1	7.5	33.9
Dose reduction	10.7	7.5	8.1	5.0	26.4
Discontinuation	28.6	15.9	18.5	5.0	16.8
Hospitalization	0	1.9	1.5	0	1.4
Duration of neratinib treatment, months					
Median	9.7	7.4	7.5	1.8	11.6
Range	0.1–13.1	0.1–12.8	0.1–13.1	0.1–6.3	0.03–13.3

[a]Non-compliance with loperamide prophylaxis in patients with grade 3 diarrhea was 71% with the original loperamide schedule, 35% with the modified loperamide schedule, and 0% with loperamide prophylaxis plus budesonide.
[b]No grade 4 events in the CONTROL study; one grade 4 event in the ExteNET study.

- There were also marked reductions in the median cumulative duration of diarrhea and in the median number of diarrhea episodes per patient with loperamide prophylaxis given with or without budesonide *vs* ExteNET (Table 2).
- The proportions of patients requiring neratinib dose holds and dose reductions were also reduced with loperamide prophylaxis given with or without budesonide *vs* ExteNET (Table 2).
- The occurrence and severity of diarrhea in the CONTROL study over the course of neratinib treatment showed a marked change from that observed in the ExteNET study (Figure 2).

Figure 2. Treatment-emergent diarrhea by month: CONTROL vs ExteNET[2]

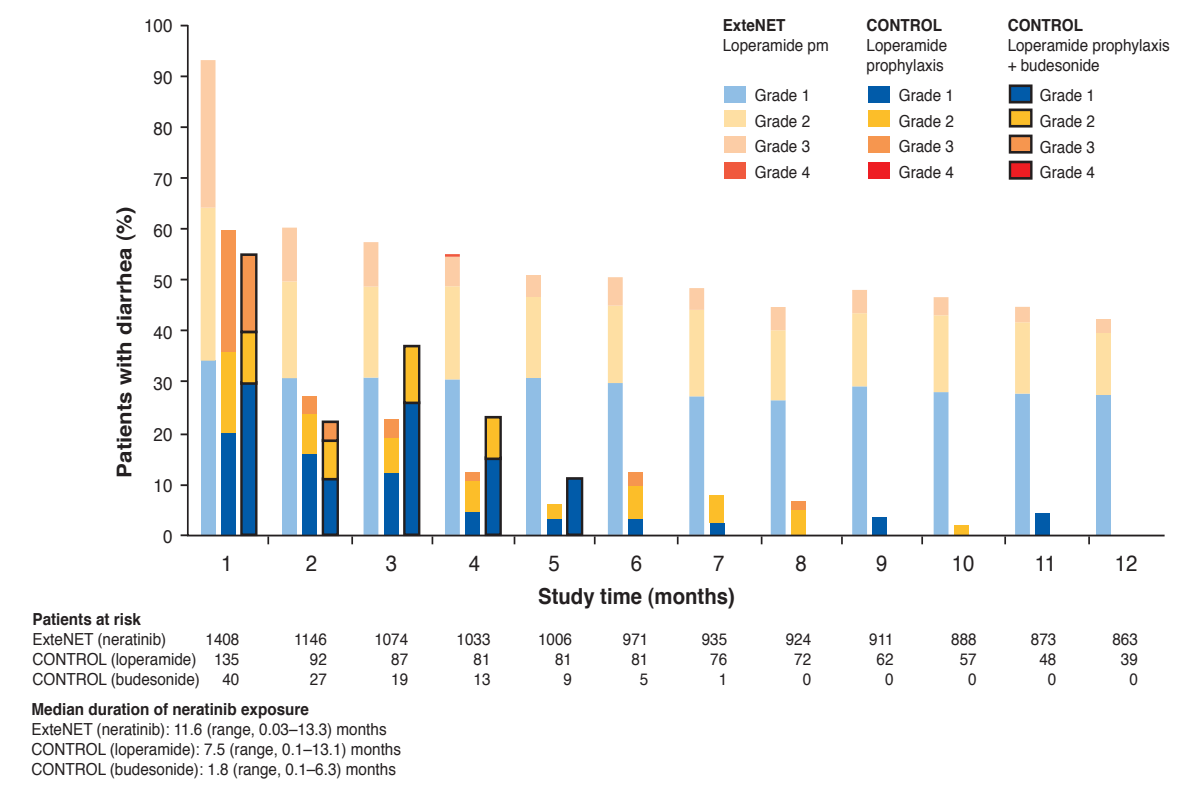
ExteNET
Loperamide prn
CONTROL
Loperamide prophylaxis
CONTROL
Loperamide prophylaxis + budesonide
Grade 1
Grade 2
Grade 3
Grade 4
Patients with diarrhea (%)
100
90
80
70
60
50
40
30
20
10
0
1 2 3 4 5 6 7 8 9 10 11 12
Study time (months)
Patients at risk
ExteNET (neratinib) 1408 1146 1074 1033 1006 971 935 924 911 888 873 863
CONTROL (loperamide) 135 92 87 81 81 81 76 72 62 57 48 39
CONTROL (budesonide) 40 27 19 13 9 5 1 0 0 0 0 0
Median duration of neratinib exposure
ExteNET (neratinib): 11.6 (range, 0.03–13.3) months
CONTROL (loperamide): 7.5 (range, 0.1–13.1) months
CONTROL (budesonide): 1.8 (range, 0.1–6.3) months

Potential factors contributing to diarrhea

- Grade 3 diarrhea rates seen in the loperamide cohort have increased over what was previously reported in December 2015 [grade 3 diarrhea, 16%, n=50 (data cut-off: September 2015)].
- Adherence to loperamide prophylaxis was assessed based on total daily dose of loperamide recorded over the first 2 cycles of neratinib treatment. Non-compliance rates with loperamide prophylaxis in patients with grade 3 diarrhea were 71% (original loperamide schedule), 35% (modified loperamide schedule), and 0% (loperamide prophylaxis plus budesonide).
- During the course of the CONTROL study, there has been an increase in the proportion of patients previously treated with pertuzumab in the neoadjuvant and adjuvant settings:
 - In the loperamide cohort, 18% of patients had previously received pertuzumab (September 2015 data cut-off) increasing to 40% (November 2016 data cut-off).
 - In the budesonide cohort, for which enrolment started most recently, 55% of patients had previously received pertuzumab.
- Table 3 shows an exploratory analysis of the incidence of grade 3 diarrhea by previous pertuzumab exposure.
 - In the loperamide cohort, grade 3 diarrhea in pertuzumab-naïve patients (23.5%) was numerically lower than in patients previously treated with pertuzumab (35.2%).
 - This may suggest that previous treatment with pertuzumab results in a higher incidence of grade 3 diarrhea with neratinib that is not able to be prevented/ reduced with loperamide prophylaxis alone.
 - Early results suggest that adding budesonide to loperamide prophylaxis may reduce grade 3 diarrhea both in patients previously treated (13.6%) and not previously treated with pertuzumab (16.7%).

Table 3. Incidence of grade 3 diarrhea by pertuzumab treatment status

	Prior pertuzumab treatment			
	Loperamide cohort		Budesonide cohort	
	Yes (n=54)	No (n=81)	Yes (n=22)	No (n=18)
Grade 3 diarrhea, %	35.2	23.5	13.6	16.7

Other adverse events

- Aside from diarrhea, the overall tolerability profile of neratinib with loperamide prophylaxis given with or without budesonide was similar to that reported in the ExteNET trial, with the exception of an increase in grade 1/2 constipation (Table 4).
- The incidence of other gastrointestinal events, i.e. nausea, vomiting, and abdominal pain, was similar in the CONTROL cohorts *vs* ExteNET.
- Grade 4 events were rare (sepsis, n=1; urinary tract infection, n=1; both unrelated events in same patient); there were no fatal adverse events in the CONTROL study.

Table 4. Other common treatment-emergent adverse events (≥10% in either CONTROL cohort)

	CONTROL				ExteNET[2]	
	Loperamide cohort		Budesonide cohort		Neratinib arm	
	Loperamide (N=135)		Loperamide + budesonide (N=40)		Loperamide prn (N=1408)	
Adverse event, %	All-grade	Grade 3/4	All-grade	Grade 3/4	All-grade	Grade 3/4
Constipation	54.1	0	47.5	0	8.2	0
Nausea	55.6	0.7	40.0	0	43.0	1.8
Fatigue	52.6	3.7	30.0	7.5	27.1	1.6
Vomiting	24.4	1.5	20.0	5.0	26.2	3.3
Abdominal pain	24.4	1.5	20.0	0	24.1	1.7
Decreased appetite	18.5	0	17.5	0	12.1	0.2
Headache	18.5	0	7.5	0	19.7	0.6
Abdominal distension	14.8	0	5.0	0	5.2	0.3
Dizziness	14.1	0	5.0	0	10.4	0.2
Dry mouth	11.9	0	5.0	0	3.3	0.1
Dehydration	5.9	1.5	10.0	2.5	3.6	0.9
Dyspepsia	6.7	0	10.0	0	9.9	0.4
Rash	3.7	0	10.0	0	15.0	0.4

Conclusions

- A structured loperamide prophylactic regimen for 2 cycles reduces the incidence, severity and duration of neratinib-associated diarrhea compared with events observed in the ExteNET trial.
- Preliminary data suggest that adding budesonide to loperamide prophylaxis may further diminish the duration and number of episodes of diarrhea, as well as decreasing the number of neratinib dose holds, dose reductions and discontinuations.
- ExteNET showed a profile for diarrhea that was chronic and characterized by higher-grade diarrhea (grades 2 and 3) that was highest in month 1 and still present in a larger proportion of patients in months 2-12. In the CONTROL study cohorts, diarrhea was characterized by a lower percentage of high-grade diarrhea in month 1 and a much lower incidence in months 2-12.
- There appears to be some adaptation to the effects of neratinib, as higher-grade diarrhea occurs early and does not typically recur.
- By controlling early diarrheal events, loperamide prophylaxis may help to improve long-term adherence and help to ensure that the efficacy benefits of neratinib are realized.
- Enrollment into the budesonide cohort is ongoing, with testing of additional investigational agents planned.
- The final analysis of the CONTROL study will be performed when all patients have completed 12 months of neratinib therapy.

References

1. Rabindran SK. *Cancer Lett* 2005;227:9–23.
2. Chan A, et al. *Lancet Oncol* 2016;17:367–77.
3. Ustaris F, et al. *Am J Hematol/Oncol* 2015;11:13–22.
4. Puma Biotechnology Inc., data on file.

Acknowledgements

CONTROL was sponsored by Puma Biotechnology Inc. Puma Biotechnology Inc. also funded the provision of editorial support provided by Miller Medical Communications.

Copyright 2016 Puma Biotechnology

Presented at the San Antonio Breast Cancer Symposium, December 6–10, 2016, San Antonio, Texas, USA
This presentation is the intellectual property of Dr. Carlos Barcenas. Contact: CHBarcenas@mdanderson.org for permission to reprint and/or distribute